Safety and Tolerability of Dimebon for up to 26 Weeks in Patients with Mild-to-Moderate Alzheimer's Disease

Mary Sano[1], Scot Styren[2], Terence Fullerton[2], Robert Brunell[2], Sarah Dubrava[2], Tom McRae[3], on behalf of the B1451027 Study Investigators
[1]Mount Sinai School of Medicine, New York; USA; [2] Pfizer Inc, New London USA; [3] Pfizer Inc, New York, USA

Introduction

- Dimebon is an orally-available investigational agent with a novel mechanism of action that has therapeutic potential in neurodegenerative diseases such as AD
- In preclinical studies, dimebon has been shown to improve mitochondrial function, protect neurons in the setting of cellular stress and enhance neuronal survival
- In a 26-week clinical study in mild-to-moderate AD subjects, dimebon demonstrated efficacy across a variety of clinical endpoints and was well tolerated
- The safety and tolerability of dimebon has been evaluated in 24 completed clinical studies in more than 2,100 subjects. Of the 1,325 subjects receiving dimebon treatment in these studies 959 were patients with AD
- The Phase 3 study (B1451027) reported here was conducted to support a mild-to-moderate AD indication for dimebon by providing additional safety data in the target population, including patients on a variety of background anti-dementia medications

Objective

- To determine the safety and tolerability of dimebon in patients with mild-to-moderate AD who were receiving either stable background anti-dementia therapy or who were not currently on anti-dementia therapy

Methods

- This was a Phase 3, multi-center, randomized, double-blind, placebo-controlled study, conducted at 102 sites in the US and Canada to evaluate the safety and tolerability of dimebon for up to 26 weeks in AD subjects (MMSE 12-26 inclusive). A total of 741 subjects were randomized into two cohorts
 - Cohort 1: 26 weeks, N = 254*
 - Cohort 2: 12 weeks, N = 487
 - Subjects were randomized 1:1 to either dimebon 10 mg or matching placebo three times daily (t.i.d.) for 7 days and then dose escalated to dimebon 20 mg or matching placebo t.i.d. for the duration of study

 *One subject in Cohort 1 was randomized but not treated.

- Key Inclusion / Exclusion criteria included:
 - Males or females ≥50 years of age
 - Diagnosis of probable AD
 - MMSE score 12 to 26, inclusive
 - Modified Hachinski Ischemic Score ≤4
 - Either on stable anti-dementia therapy or not on anti-dementia therapy for 60 days prior to study start
- Safety and tolerability was assessed at each visit by recording of adverse events (AEs) and monitoring of vital signs, physical examinations, safety laboratory evaluations, and ECGs

Results

- Demographic characteristics were generally comparable across the treatment groups for both Cohort 1 and 2 (Table 1 shows the total study data.)

Table 1: Subject Demographics

Subject Demographics	Dimebon	Placebo
Number (%) of subjects		
Number of subjects	370	371
Sex		
Male	178 (48.1)	173 (46.6)
Female	192 (51.9)	198 (53.4)
Age (years)		
Mean (SD)	75.8 (8.7)	76.0 (8.2)
Range	50 -94	50-99
Years since AD diagnosis		
Mean	2.9	2.9
Range	0 -12.4	0 -12.5
MMSE score		
Mean (SD)	20.6 (4.0)	20.2 (4.1)
Range	12-26	12-17
CYP2D6 metabolizer status		
PM	16 (4.3)	19 (5.1)
IM	12 (3.2)	15 (4.0)
EM	297 (80.3)	276 (74.4)
UM	7 (1.9)	15 (4.0)
ApoE genotype		
E4+ (at least 1 E4allele)	229 (61.9)	228 (61.5)
E4 - (no E4 allele)	135 (36.5)	136 (36.7)

- Most subjects were on background anti-dementia therapy (Table 2)

Table 2: Use of Background Anti-Dementia Therapy

Number (%) of subjects	Dimebon N = 370	Placebo N = 371
Any anti-dementia therapy	303 (82)	315 (85)
Cholinesterase inhibitors	152 (41)	158 (43)
Memantine	20 (5)	24 (6)
Combination of above	131 (35)	133 (36)
None	67 (18)	56 (15)

- Treatment compliance was high in both the dimebon and placebo groups (80 % overall compliance)
 - Dimebon-treated subjects: 341 (92.2 %)
 - Placebo-treated subjects: 331 (89.2 %)
- Most treatment-emergent AEs were mild or moderate
- The incidence of treatment-emergent AEs was similar between the dimebon and placebo groups overall (Table 3) and within each cohort
 - Consistent with the longer duration, there was a slightly higher percentage of subjects with AEs in Cohort 1(dimebon 64.6 %, placebo 64.6 %) than in Cohort 2 (dimebon 55.1 %, placebo 53.3 %) but no clinically important treatment difference in either cohort

Table 3: Treatment-Emergent Adverse Events, All Causalities

Number (%) of subjects	Overall	
	Dimebon	Placebo
Subjects evaluable for AEs	370	371
Number of AEs	536	531
Subjects with AEs	216 (58.4)	212 (57.1)
Subjects with SAEs	26 (7.0)	28 (7.5)
Subjects with severe AEs	23 (6.2)	21 (5.7)
Subjects discontinued due to AEs	21 (5.7)	18 (4.9)
Subjects with dose reduced or temporary discontinuation due to AEs	16 (4.3)	22 (5.9)

- Urinary tract infection was the most commonly reported treatment-emergent AE (Table 4)
 - Fatigue and somnolence were the only AEs that occurred in the dimebon group at an incidence of ≥2 % and twice the placebo group [fatigue (5.1 % vs. 2.4 %); somnolence (5.1 % vs. 1.9 %)]

Table 4: Incidence of Treatment-Emergent Adverse Events by System Organ Class and Preferred Term in 2% or More Subjects Overall in Either Treatment Group, All Causalities

System Organ Class Preferred Term, n (%)	Dimebon N = 370	Placebo N = 371
Nervous system disorders	**66 (17.8)**	**61 (16.4)**
Somnolence*	19 (5.1)	7 (1.9)
Headache	14 (3.8)	10 (2.7)
Dizziness	9 (2.4)	9 (2.4)
Infections and infestations	**58 (15.7)**	**52 (14.0)**
Urinary tract infection	27 (7.3)	28 (7.5)
General disorders and administration site conditions	**49 (13.2)**	**29 (7.8)**
Fatigue*	19 (5.1)	9 (2.4)
Oedema peripheral	11 (3.0)	9 (2.4)
Gastrointestinal disorders	**44 (11.9)**	**54 (14.6)**
Diarrhoea	13 (3.5)	9 (2.4)
Nausea	8 (2.2)	9 (2.4)
Constipation[ψ]	2 (0.5)	9 (2.4)
Psychiatric disorders	**41 (11.1)**	**49 (13.2)**
Agitation	8 (2.2)	5 (1.3)
Anxiety[ψ]	3 (0.8)	9 (2.4)
Confusional state[ψ]	4 (1.1)	10 (2.7)
Insomnia	5 (1.4)	9 (2.4)
Injury, poisoning and procedural complications	**31 (8.4)**	**30 (8.1)**
Fall	15 (4.1)	19 (5.1)
Musculoskeletal and connective tissue disorders	**24 (6.5)**	**26 (7.0)**
Back pain	5 (1.4)	10 (2.7)

*more than twice placebo [ψ]more than twice dimebon

- The incidence of SAE's was similar between subjects treated with dimebon or placebo (7.0 % vs 7.5 %) (Table 3)
- Three deaths were reported during the B1451027 study, with only 1 death reported in the dimebon-treated group (secondary to complications of recurrent T-cell lymphoma)

Subgroup Analyses

- The incidence of AEs was similar between subjects receiving anti-dementia therapy and subjects not receiving anti-dementia therapy and between dimebon and placebo within these subgroups (Table 5)

Table 5: Treatment-Emergent Adverse Events by Background Anti-Dementia Therapy Status, All Causalities

Number (%) of subjects	Receiving Any Anti-Dementia Therapy		Not Receiving Any Anti-Dementia Therapy	
	Dimebon[Ω]	Placebo[α]	Dimebon[δ]	Placebo[Φ]
Overall				
Subjects evaluable for AEs	303	315	67	56
Number of AEs	453	438	83	93
Subjects with AEs	176 (58.1)	178 (56.5)	40 (59.7)	34 (60.7)
Subjects with SAEs	21 (6.9)	27 (8.6)	5 (7.5)	1 (1.8)
Subjects with severe AEs	20 (6.6)	16 (5.1)	3 (4.5)	5 (8.9)
Subjects discont' due to AEs	19 (6.3)	15 (4.8)	2 (3.0)	3 (5.4)
Subjects dose reduced or temp discontinuation due to AEs	13 (4.3)	16 (5.1)	3 (4.5)	6 (10.7)

[Ω]Mean Age: 75.6y, Mean MMSE 20.4 / [α]Mean Age: 75.9y, Mean MMSE: 20.8
[δ]Mean Age: 76.5y, Mean MMSE: 21.9 / [Φ]Mean Age: 76.5y, Mean MMSE: 21.0

- Dimebon is metabolized via the CYP2D6 pathway, such that PMs* are expected to have significantly higher drug exposure than EMs*. When examined by CYP2D6 metabolizer status, the incidence of AEs was similar among UMs*, EMs, IMs*, and PMs and between dimebon and placebo within these subgroups (Figure 1)
 - AEs were also similar among subgroups of subjects not receiving moderate/potent CYP2D6 inhibitors, receiving moderate CYP2D6 inhibitors, and receiving potent CYP2D6 inhibitors
 - Small sample sizes in many categories limit the interpretation

*UM; ultra metabolizer, EM: extensive metabolizer, IM: intermediate etabolizer, PM: poor metabolizer

Figure 1. % of Subjects with AEs by CYP2D6 Metabolizer Status and Treatment

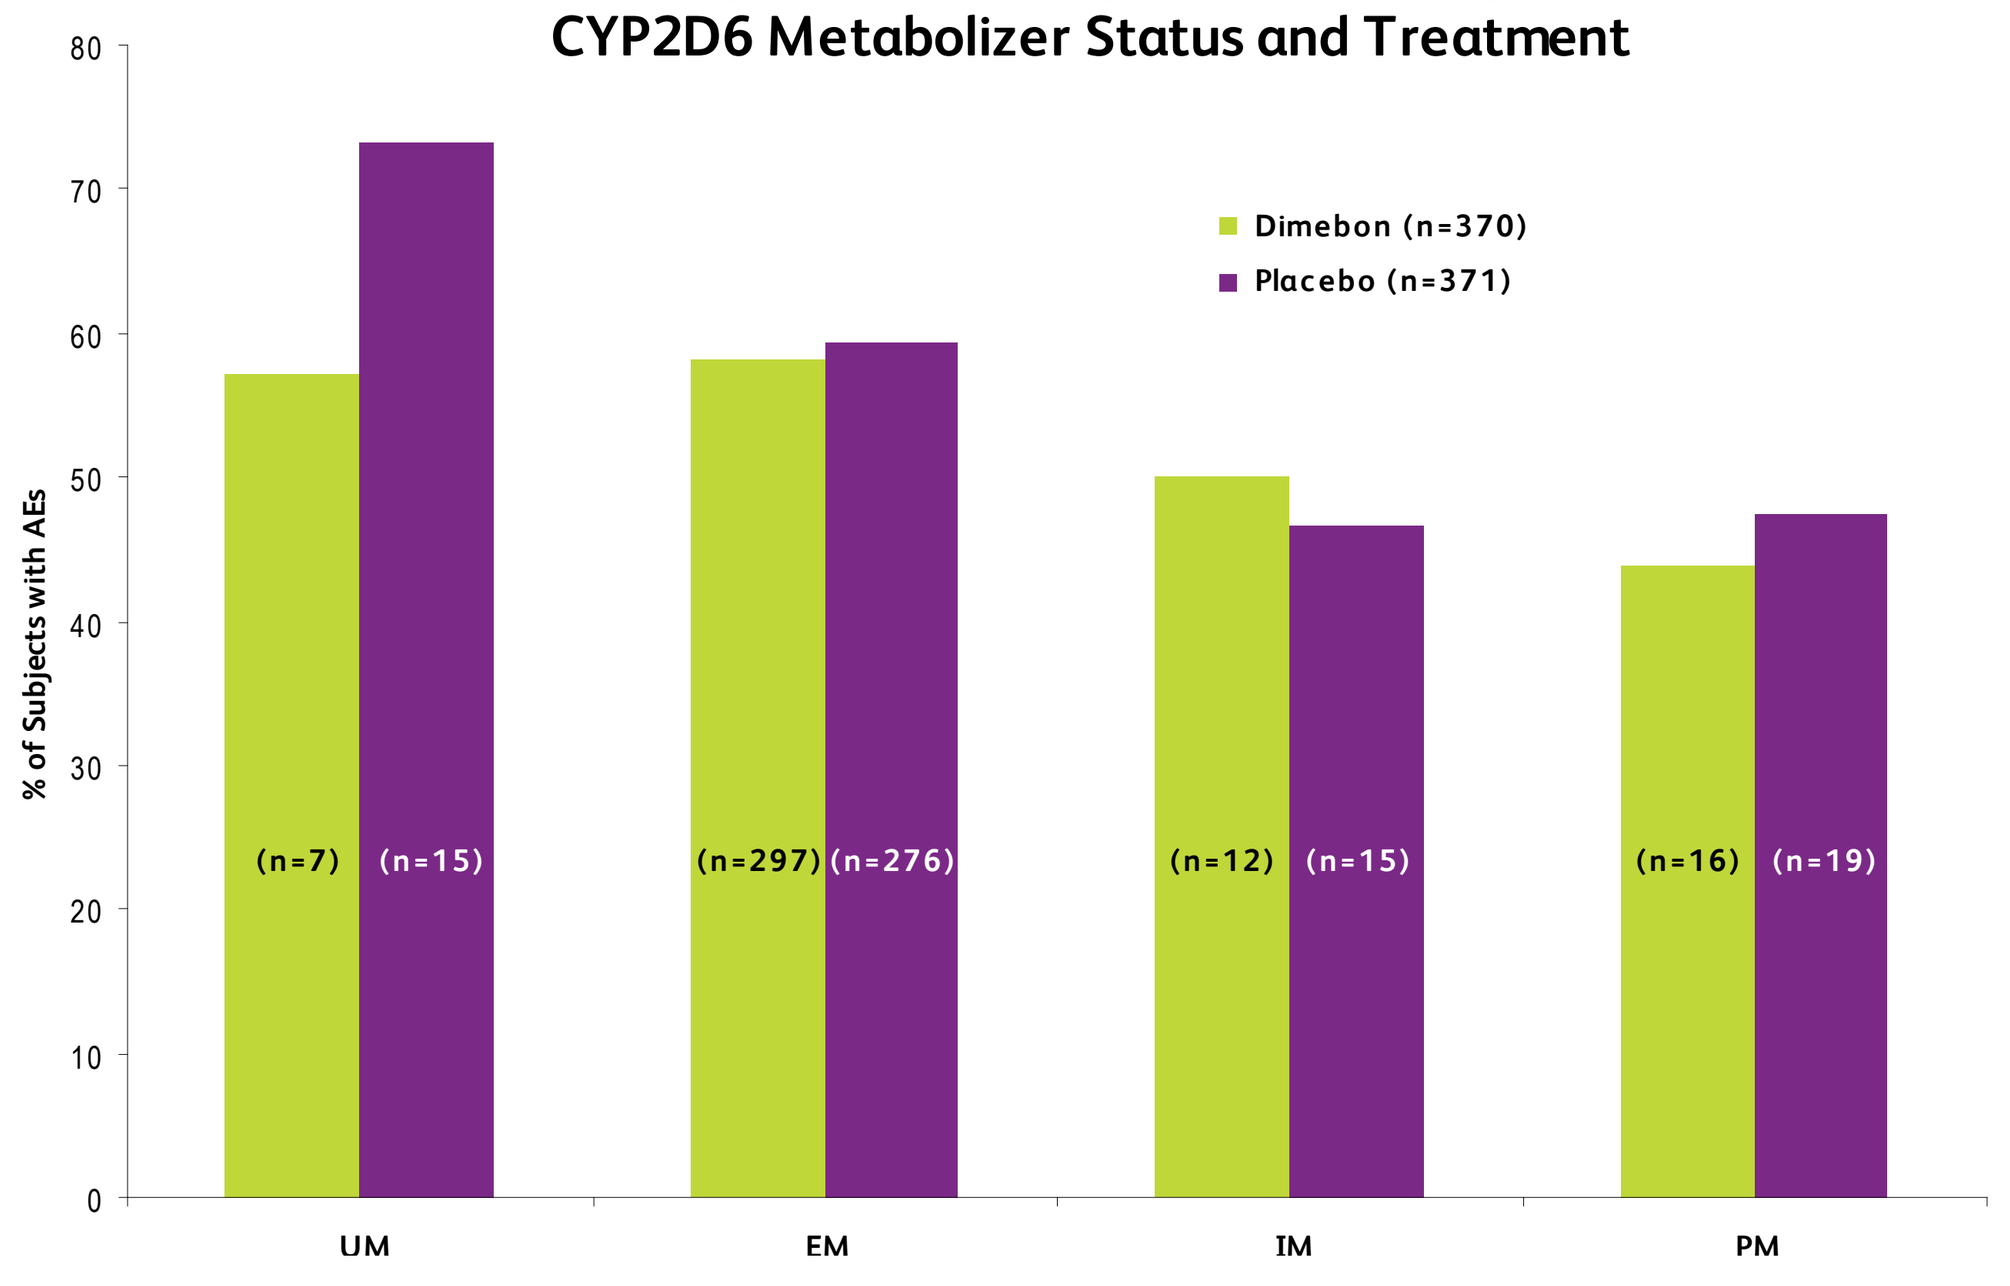
Dimebon (n=370)
Placebo (n=371)
% of Subjects with AEs
(n=7) (n=15)
(n=297) (n=276)
(n=12) (n=15)
(n=16) (n=19)
UM
EM
IM
PM

*UM; ultra metabolizer, EM: extensive metabolizer, IM: intermediate metabolizer, PM: poor metabolizer

Figure 2. % of Subjects with AEs by Treatment and by Background Treatment with Moderate or Potent CYP2D6 Inhibitors and by Background Psychotropic Use

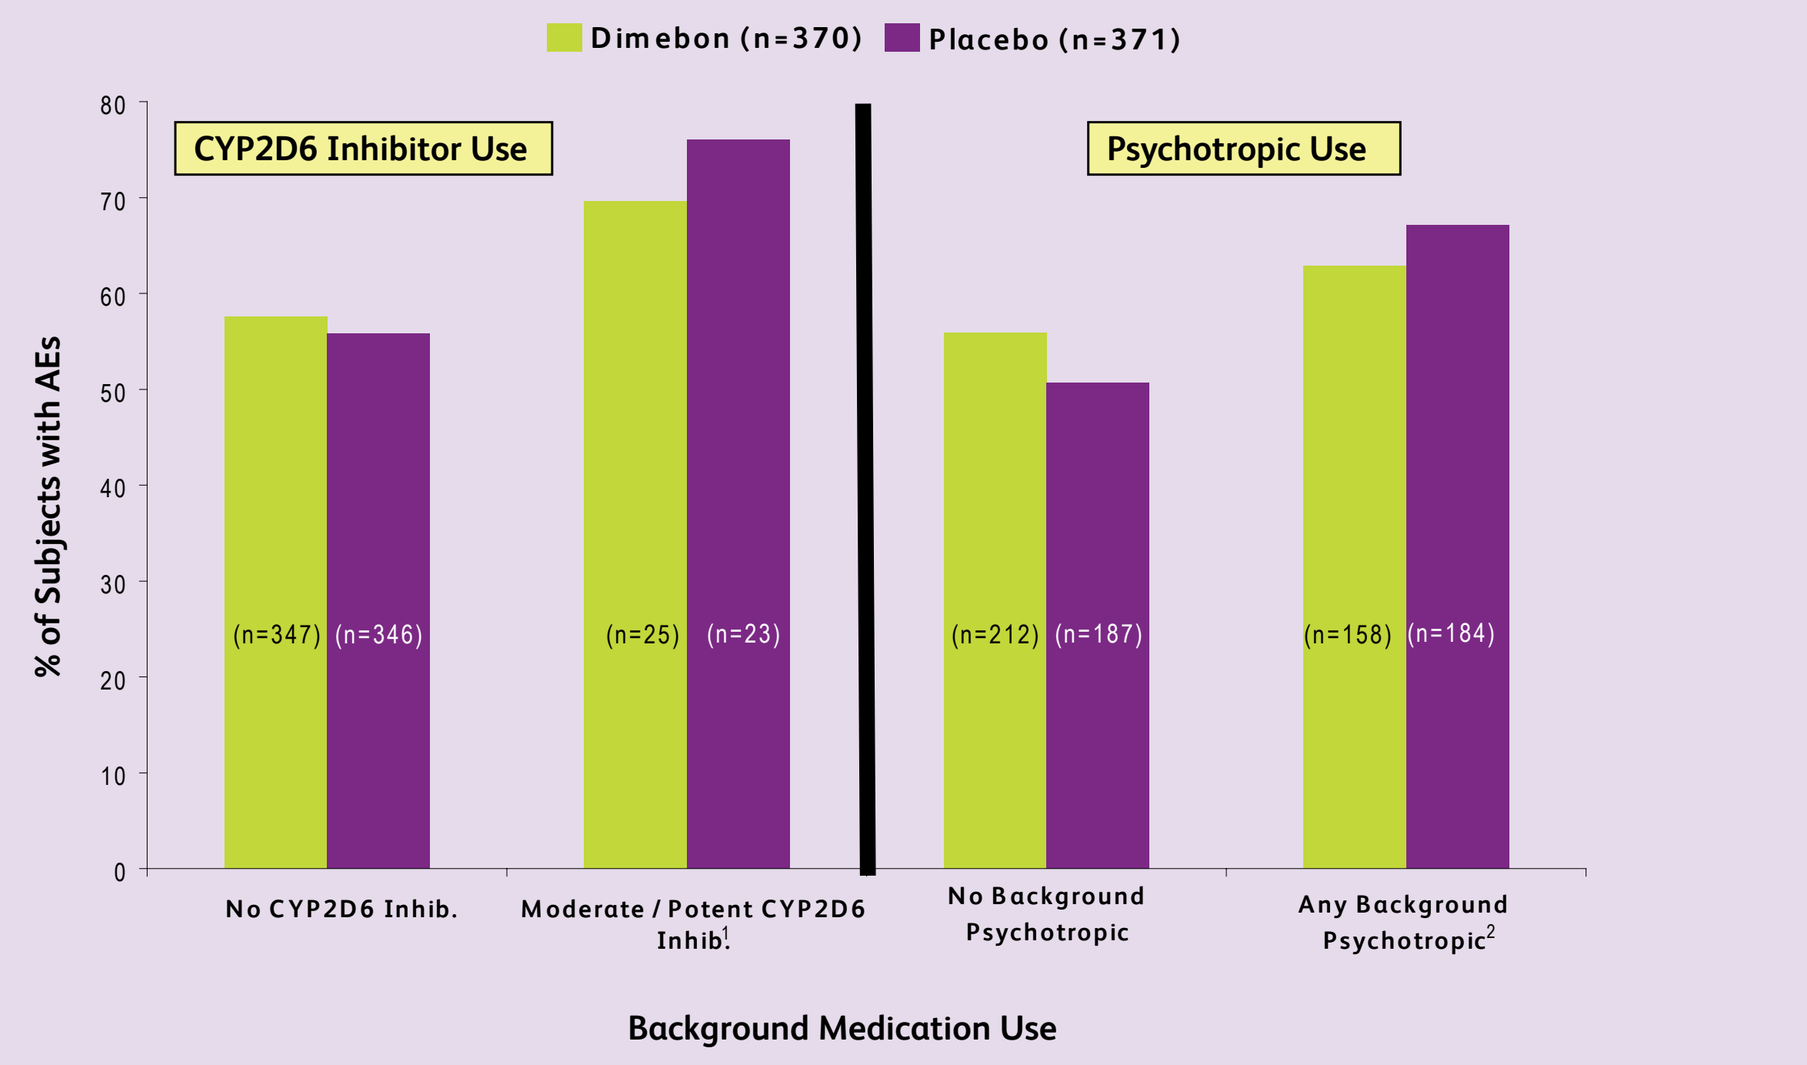
Dimebon (n=370)
Placebo (n=371)
CYP2D6 Inhibitor Use
Psychotropic Use
% of Subjects with AEs
(n=347) (n=346)
(n=25) (n=23)
(n=212) (n=187)
(n=158) (n=184)
No CYP2D6 Inhib.
Moderate / Potent CYP2D6 Inhib.[1]
No Background Psychotropic
Any Background Psychotropic[2]
Background Medication Use

[1]Potent inhibitor was defined as paroxetine, fluoxetine, or quinidine. Moderate inhibitor was defined as duloxetine or terbinafine.
[2] Sedatives, hypnotics anxiolytics, anti-depressants, anti-psychotics

- Analysis of APOE (4) carrier status in subjects randomized to dimebon revealed that the incidence of AEs was somewhat higher in those who were E4– (65.9 %) compared to subjects who were E4+ (53.7 %). However, no consistent or meaningful differences were observed between dimebon and placebo within each of these subgroups

Analysis of Other Safety Endpoints

- No clinically important differences were seen in:
 - MMSE scores at 12 weeks (dimebon: +0.04 points, Placebo: +0.11 points)
 - Clinical laboratory abnormalities
 - Vital signs or ECG parameters
- A higher incidence of positive urine Hgb was observed in the dimebon treatment group vs. placebo (28 % vs. 6 %) but was not associated with an increase in RBCs on microscopic exam, urine protein or creatinine clearance, suggesting a false-positive dipstick assay (Figure 3)

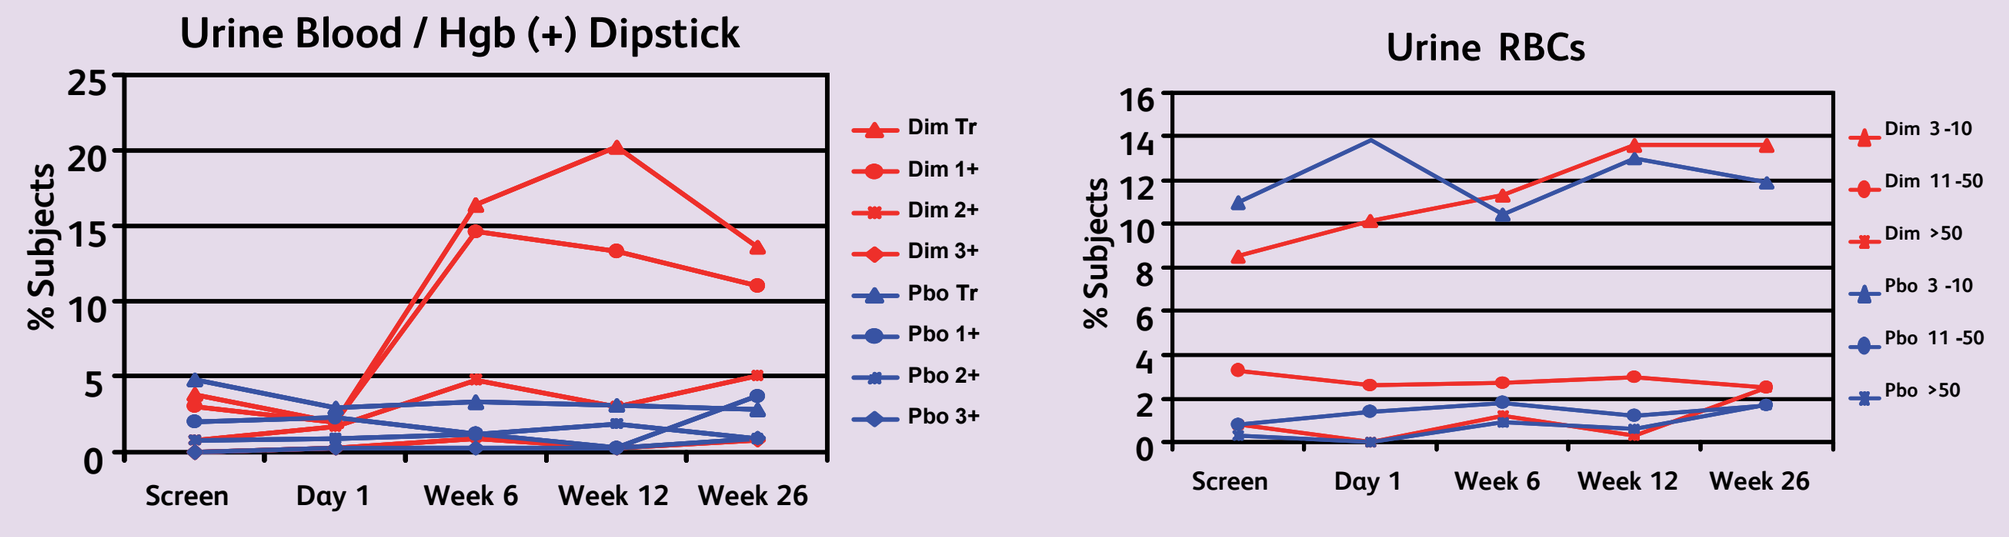
Urine Blood / Hgb (+) Dipstick
% Subjects
Screen
Day 1
Week 6
Week 12
Week 26
Dim Tr
Dim 1+
Dim 2+
Dim 3+
Pbo Tr
Pbo 1+
Pbo 2+
Pbo 3+
Urine RBCs
% Subjects
Dim 3-10
Dim 11-50
Dim >50
Pbo 3-10
Pbo 11-50
Pbo >50

Conclusions

- In this study, dimebon treatment at a dose of 20 mg three times daily for up to 26 weeks was well tolerated in subjects with mild-to-moderate AD on a variety of background anti-dementia medications
- There was no notable influence of any demographic or clinical feature on the overall assessment of the safety and tolerability of dimebon in this study

Poster presented at the ICAD 2010 Annual Meeting; July 10–15, 2010; Honolulu, Hawaii, USA