A Multicenter, Open-label, Proof of Concept Phase 1b/2 Study to Evaluate the Safety and Efficacy of Archexin® (RX-0201) in Combination With Everolimus to Treat Subjects With Advanced Renal Cell Carcinoma.

The Rexahn Investigator Study Team[1], Christine Peterson[2], and Ely Benaim[2]

[1] *Various Study Centers*; [2] Rexahn Pharmaceuticals, Inc. Rockville, MD, USA.

Abstract #TPS4580

Background: RX-0201 is a 20-mer oligonucleotide that is complementary to AKT-1 messenger ribonucleic acid (mRNA). The specificity of RX-0201-mediated effect on AKT-1 mRNA levels was examined in human renal cell carcinoma (von Hippel-Lindau protein-deficient renal cell carcinoma cell line) UMRC2 cells and resulted in a reduction of AKT-1 mRNA levels.

Methods: The current study is a proof of concept phase 1b/2, multicenter, open label study conducted in 2 stages. Stage 1 is an open-label, dose-escalation phase 1b study of Archexin® **(**RX-0201) administered in combination with everolimus. RX-0201 will be administered by a 24 hour continuous intravenous infusion for 14 days followed by 7 days of rest. It is expected that 250 mg/m^2/day or a lower dose of RX-0201 will be identified as safe and well-tolerated when administered in combination with 10 mg of everolimus. The dose of RX-0201 identified in Stage 1 will be studied further in Stage 2 which is the randomized, open-label, 2-arm study of RX-0201 in combination with 10 mg of everolimus versus 10 mg of everolimus alone. Up to 8 cycles of study treatment will be permitted. Approximately 9 subjects are targeted to receive escalating doses of RX-0201 in combination with everolimus in Stage 1. The initial dose of RX-0201 is 125 mg/m^2/day. In Stage 2 approximately 30 subjects are planned to be randomized in a 1:2 ratio (i.e., up to 10 subjects in the everolimus arm and up to 20 subjects in the everolimus/ RX-0201 arm). Eligible subjects must have confirmed histologic or cytologic evidence of renal cancer with a clear cell component, measurable or evaluable disease as defined by RECIST, received at least 1 course of therapy with a VEGFR inhibitor and progressed within 6 months of planned first dose of on study treatment and received no more than 3 prior treatments of systemic renal cancer therapy. Radiological imaging for disease assessments will be according to RECIST ver. 1.1

NCT02089334

Prior Clinical Experience

Phase 1 single agent dose escalation study of Archexin® (RX-0201) enrolled 7 subjects with solid tumors that were treated with doses ranging from 6.0 to 315 mg/m^2/day. The maximum tolerated dose of single agent RX-0201 was 250 mg/m^2/day. Fatigue was the most commonly reported adverse event in 82% of subjects (14/17). Arthralgia (joint pain) and nausea were the second most commonly reported adverse events with 47% (8/17) and 35% (6/17) of subjects reporting these events, respectively.

Phase 2 study of Archexin® and gemcitabine in metastatic pancreatic cancer was a 2 stage study. Stage 1 enrolled 11 subjects that received gemcitabine at 1000 mg/m^2 for 30 min followed by a 14 day continuous infusion of escalating doses of RX-0201 to determine the MTD.

Stage 2 enrolled 20 subjects that received gemcitabine in combination with followed by 200 mg/m^2/day of Archexin®. The most commonly reported treatment emergent adverse events related to Archexin® were thrombocytopenia (8.7%), decreased platelet count (7.7%), neutropenia (6.7%), fatigue (5.8%), and decreased hemoglobin (5.8%).

Proposed Mechanism

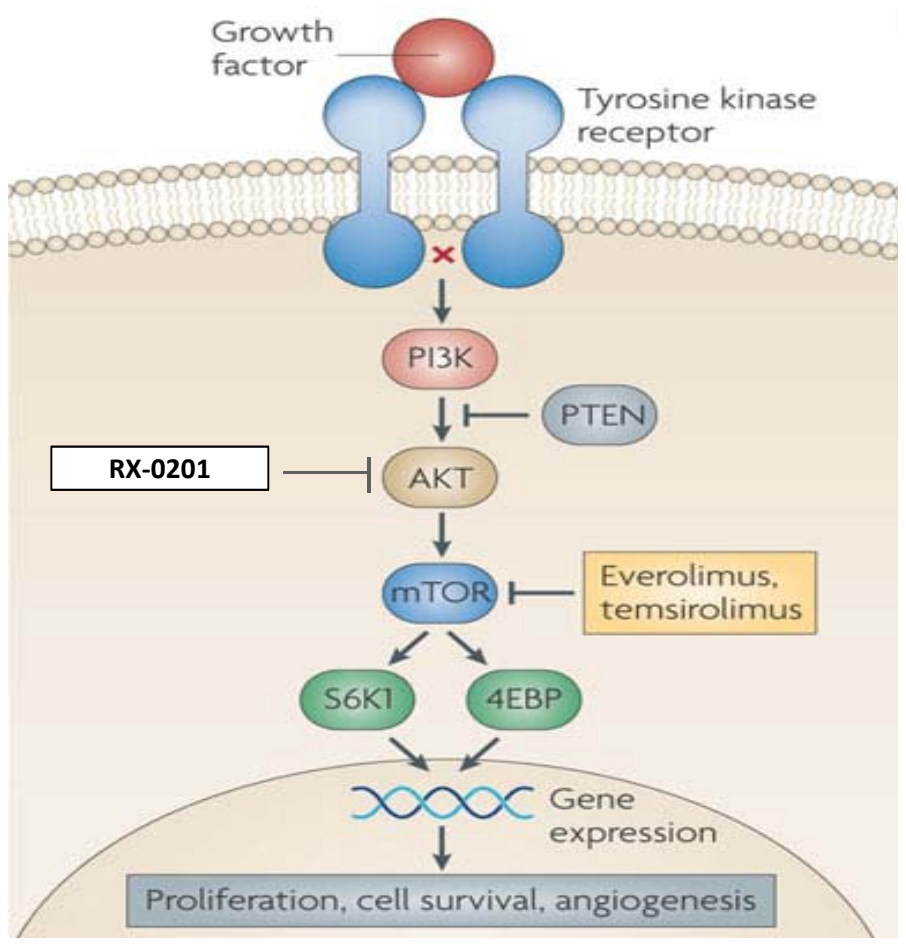
Growth factor
Tyrosine kinase receptor
PI3K
PTEN
RX-0201
AKT
mTOR
Everolimus, temsirolimus
S6K1
4EBP
Gene expression
Proliferation, cell survival, angiogenesis

Adapted from Atkins, M.B., Yasothan, U. & Kirkpatrick, P. Everolimus. *Nature Reviews Drug Discovery* 8, 535-536 (July 2009)

Study Design

Methodology: This study is a 2-stage multi-center, open-label study to assess the safety and tolerability of Archexin® (RX-0201) in combination with everolimus vs everolimus alone to treat subjects with advanced renal cell carcinoma.

Stage 1 is an open-label, dose-escalation study designed to identify a safe and tolerable dose of Archexin® (RX-0201) when given in combination with everolimus

Stage 2 is a randomized, open-label, 2-arm dose expansion study of Archexin® (RX-0201) in combination with everolimus versus everolimus alone for up to 8 cycles to determine safety and efficacy of the combination. Subjects will receive RX-0201, at the dose identified in Stage 1, in combination with everolimus or everolimus alone,. Subjects will be randomized in a 1:2 ratio (i.e., up to 10 subjects in the everolimus alone arm and up to 20 subjects in the everolimus/ RX-0201 arm).

Treatment: RX-0201 is administered by continuous IV infusion for 14 days followed by 1 week of rest.

In Stage 1 and Stage 2, everolimus (10 mg) will be administered daily according to the approved label instructions.

Dosing: The initial dose of RX-0201 is 125 mg/m^2/day in Stage 1. The dose will be escalated until the maximum tolerated dose is identified or target dose is achieved. Subsequent dose levels are 200 mg/m^2/day and 250 mg/m^2/day.

The dose of RX-0201 identified in Stage 1 will be studied further in the dose expansion portion (Stage 2).

Study Objectives

Primary Objectives:
- To determine the maximum tolerated dose (MTD) of RX-0201, up to a target dose of 250 mg/m^2/day, when given in combination with everolimus (Stage 1)
- To determine progression free survival in subjects with advanced renal cell carcinoma treated with the combination of RX-0201 and everolimus versus everolimus alone (Stage 2)

Secondary Objectives:
- To assess the pharmacokinetics of RX-0201 in combination with everolimus (Stage 1)
- To evaluate parameters of clinical benefit as measured by duration of response, time to response, and response rate (Stage 2)
- To evaluate the safety and tolerability of RX-0201 in combination with everolimus versus everolimus alone (Stage 1 and Stage 2)

Exploratory Objective:
- To investigate blood or tumor response/ resistance to RX-0201 and everolimus as assessed by AKT pathway biomarkers, tumor apoptosis biomarkers and other biomarkers (Stage 1 and Stage 2)

Study Endpoints

Primary Endpoints
- Incidence of adverse events and clinical laboratory abnormalities defined as dose-limiting toxicities (Stage 1)
- Progression free survival (Stage 2)

Secondary Endpoints
- Pharmacokinetic profile of RX-0201 (Stage 1)
- Incidence of adverse events, changes in clinical laboratory tests and vital signs over time (Stage 1 and Stage 2)
- Tumor response, duration of response, time to response, and response rates (Stage 2)

Key Inclusion Criteria

- Males and females ≥ 18 years of age at screening
- Histological or cytological diagnosis of renal cell cancer with a clear-cell component
- Measurable or evaluable disease defined by Response Evaluation Criteria for Solid Tumors (RECIST) ver. 1.1
- Must have received at least one course of therapy with a VEGFR-targeting tyrosine kinase inhibitor (eg, sorafenib, sunitinib, axitinib, pazopanib or tivozanib) and progressed within 6 months of planned first dose of study treatment
- No more than 3 prior treatments of systemic therapy for renal cancer
- ECOG performance status of 0,1 or 2

Key Exclusion Criteria

- Brain metastases or cranial epidural disease unless adequately treated with radiotherapy and/or surgery and stable for at least 3 months before planned first dose of study drug
- Radiation therapy for bone metastasis within 2 weeks, any other external radiation therapy within 4 weeks before planned first dose of study drug. Systemic treatment with radionuclides within 6 weeks before planned first dose of study drug. Subjects with clinically relevant ongoing complications from prior radiation therapy are not eligible
- Prior treatment with everolimus, or any other specific or selective TORC1/PI3K/AKT inhibitor (eg, temsirolimus)
- Receipt of any type of small molecule kinase inhibitor (including investigational kinase inhibitor) within 2 weeks before planned first dose of study drug
- Receipt of any type of anticancer antibody (including investigational antibody) within 4 weeks before planned first dose of study drug
- Taking strong inducers or inhibitors of CYP450s for subjects receiving everolimus
- Chronic treatment with corticosteroids or other immuno-suppressive agents
- Concomitant anticoagulation at therapeutic doses with oral anticoagulants or platelet inhibitors
- Subjects with a known hypersensitivity to everolimus or other rapamycins (sirolimus, temsirolimus) or to its excip-ients
- Active infection requiring parenteral antibiotics within 2 weeks before planned first dose of study drug

Investigator Disclosures

1. Christine Peterson, PhD – Rexahn Pharmaceuticals
2. Ely Benaim, MD – Rexahn Pharmaceuticals

For further information about RX-0201 and Rexahn Pharmaceuticals
please contact Dr. Christine Peterson: petersonc@rexahn.com, (240) 268-5300 x 320