A Phase 1 Study of RX-3117 an Oral Agent Activated by Uridine Cytidine Kinase 2 to Treat Subjects with Advanced Solid Tumors.

Drew Rasco[1], Amita Patnaik[1], Anthony Tolcher[1], Kyriakos Papadopoulos[1], Christine Peterson[2], and Ely Benaim[2],
[1]South Texas Accelerated Research Therapeutics, San Antonio, TX, [2]Rexahn Pharmaceuticals Inc., Rockville, MD

Abstract TPS#2605

Background: RX-3117 is an oral small-molecule antimetabolite, cyclopentyl pyrimidyl nucleoside that is activated by uridine cytidine kinase 2. RX-3117's efficacy in xenograft models (Colo-205, H460, H69 and CaSki), which are moderately sensitive or resistant to gemcitabine, indicates that RX-3117 may have the potential to treat tumors that do not respond to gemcitabine or have become gemcitabine resistant.

Methods: This Phase 1, open-label, multicenter study evaluates the efficacy and safety of RX-3117 in subjects with solid tumors. RX-3117 is administered 3 times a week for 3 weeks with 1 week off during each 4 week cycle. Dose escalation starts with an accelerated design treating 1 subject per dose followed by a standard 3 + 3 design using a modified Fibonacci sequence after the occurrence of a single related Grade 2 or greater adverse event. The primary endpoint is the overall safety profile characterized by the type, frequency, severity, timing of onset, duration and relationship to study therapy of any adverse events, or abnormalities of laboratory tests or electrocardiograms, any dose limiting toxicities that occur during Cycle 1, serious adverse events, or adverse events leading to study treatment discontinuation. Secondary endpoints include pharmacokinetic parameters (e.g., time to maximum observed concentration [T_{max}], maximum observed plasma concentration [C_{max}], trough concentration [C_{trough}], area under the concentration-time curve [AUC]) and Indices of anti-tumor activity (e.g., overall response rate , time to response, duration of response, and progression-free survival). Exploratory endpoints are baseline biomarker expression/-concentration, including (but not limited to) concentrative nucleoside transporter 2; equilibrative nucleoside transporter 1; uridine-cytidine kinase 1 and 2; DNA methyltransferase 1, 3a and 3b; and ribonucleotide reductases 1 and 2. Target recruitment is approximately 30 subjects. Eligible subjects must have confirmed histologic or cytologic evidence of metastatic or locally advanced solid neoplasm that has failed to respond to standard therapy, progressed despite standard therapy or for which standard therapy does not exist.

NCT02030067

Mechanism of Action

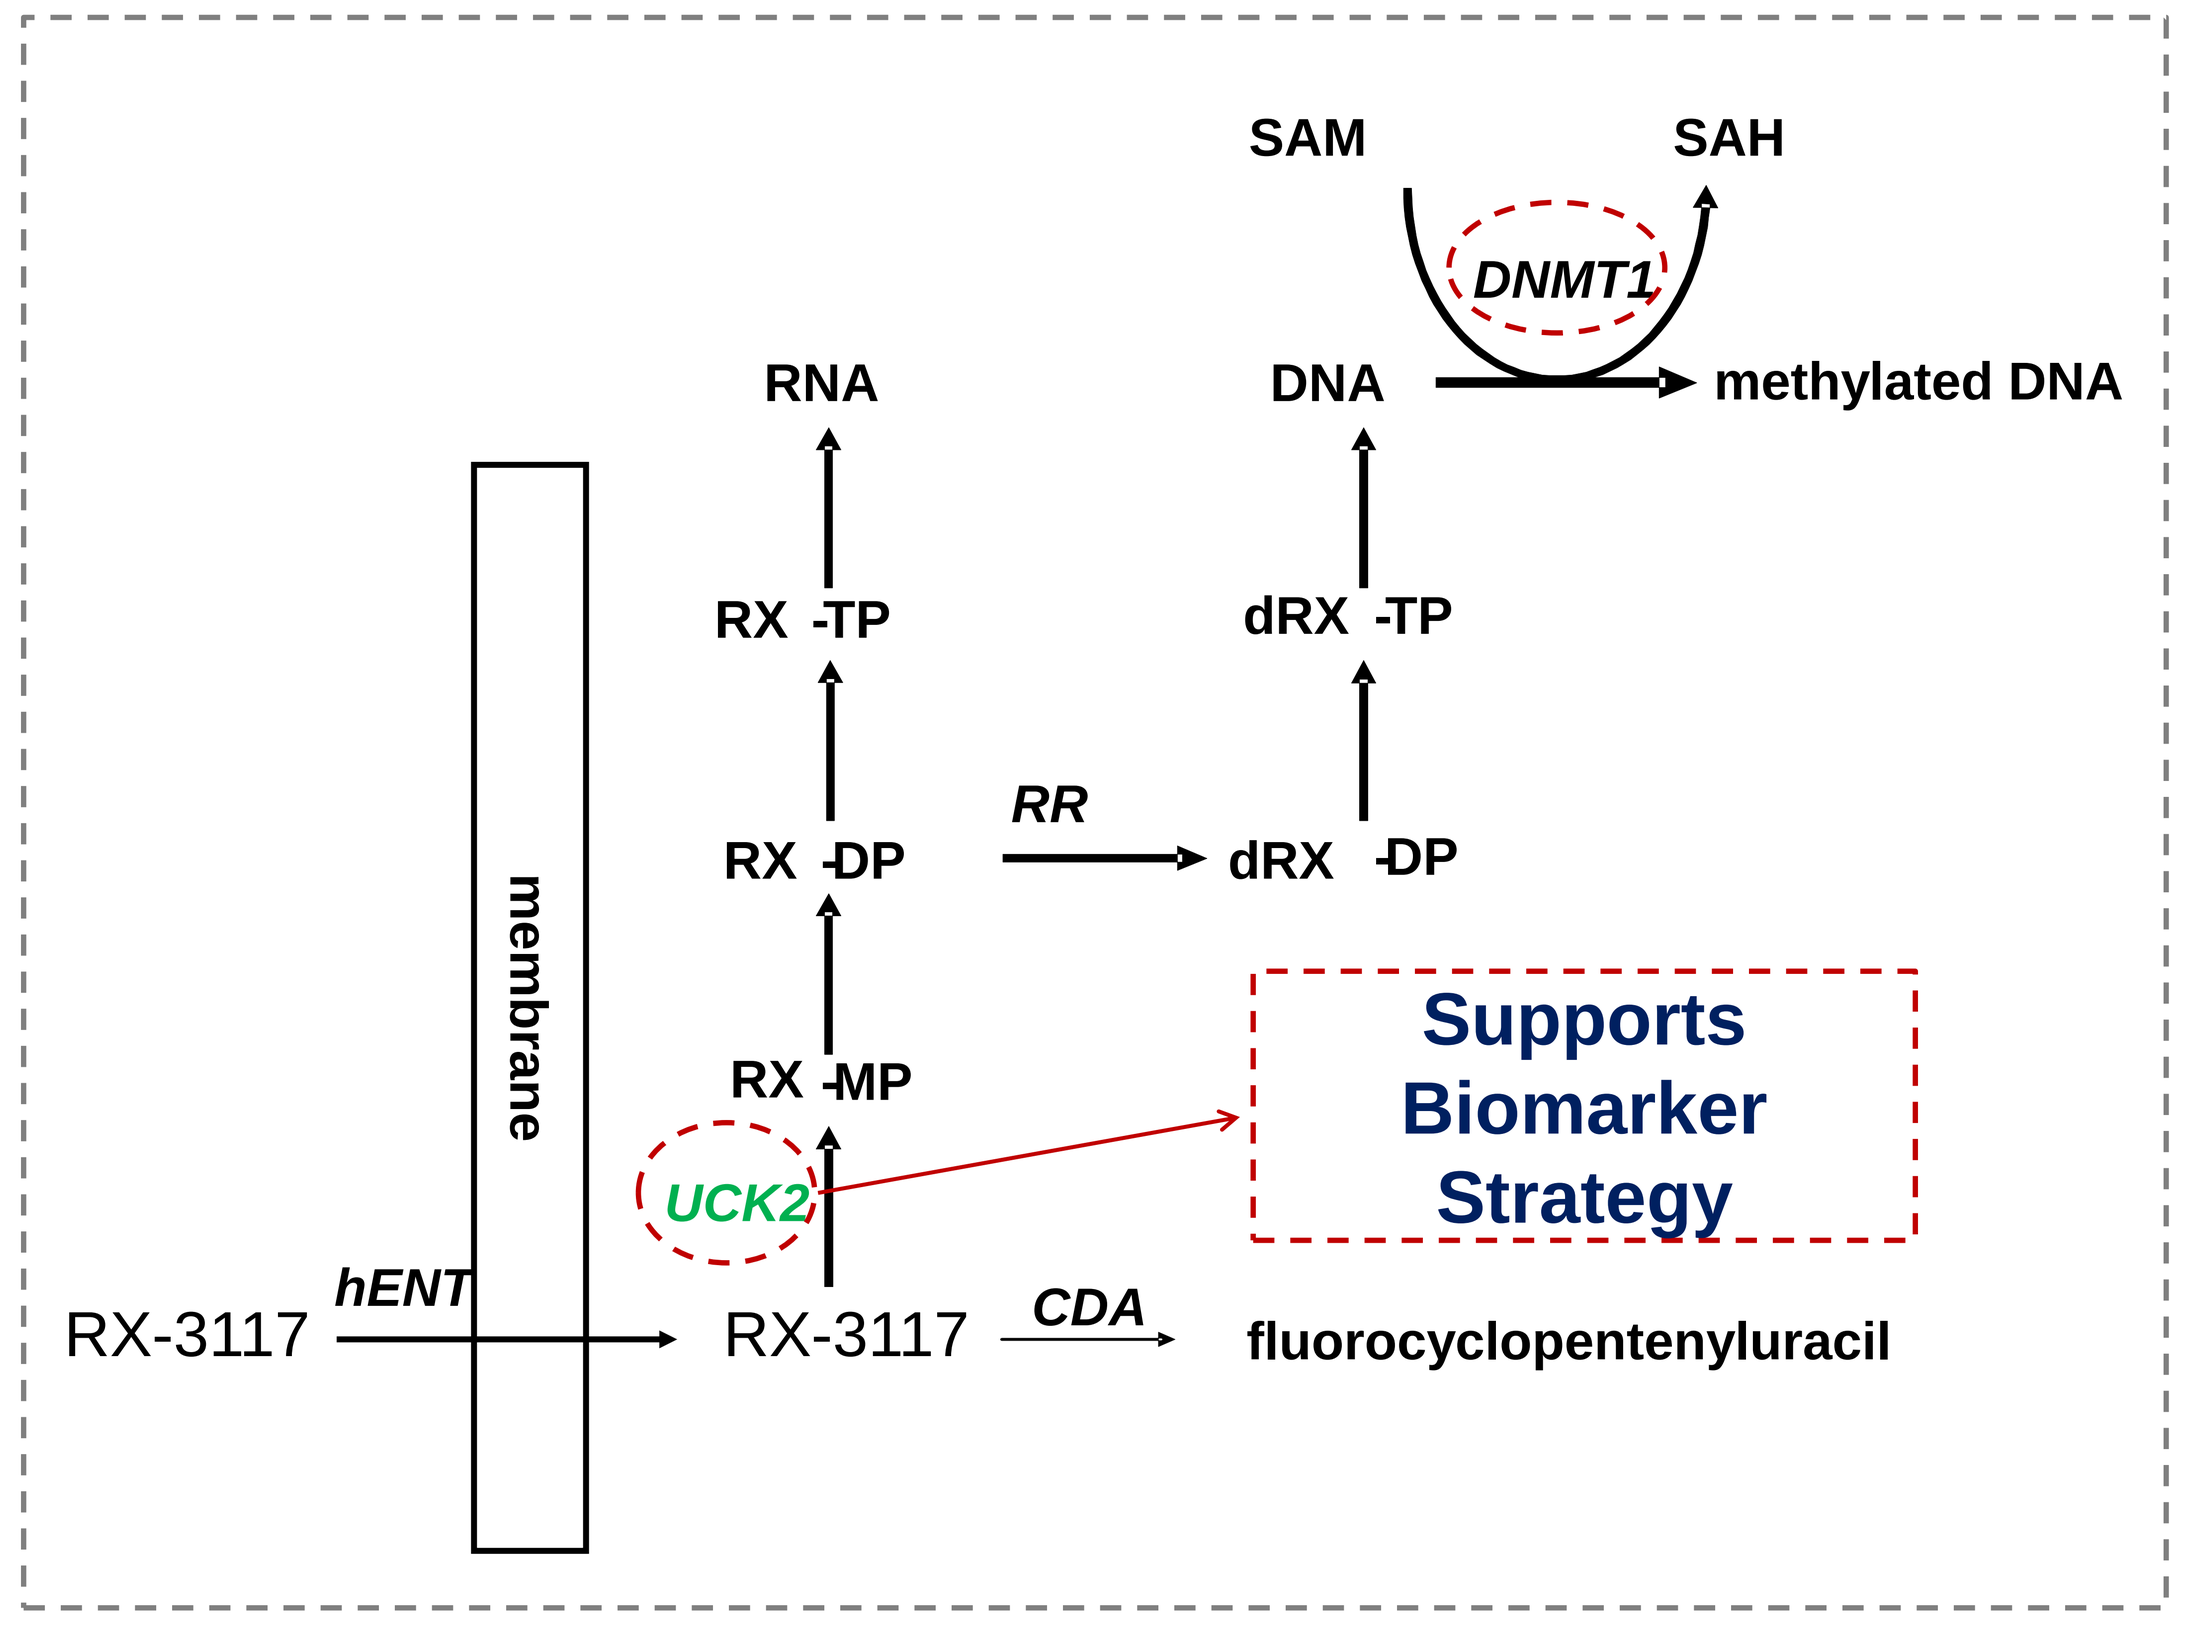
SAM
SAH
DNMT1
RNA
DNA
methylated DNA
RX -TP
dRX -TP
RR
RX -DP
dRX -DP
membrane
RX -MP
Supports Biomarker Strategy
UCK2
RX-3117
hENT
RX-3117
CDA
fluorocyclopentenyluracil

- RX-3117 is incorporated into both RNA and DNA and down regulates DNA methyltransferase (DNMT1).
- RX-3117 is degraded by cytidine deaminase more slowly than gemcitabine.
- RX-3117 is activated by uridine cytidine kinase (UCK2).
- UCK2 appears to be overly expressed in several types of cancer cells with limited presence in normally dividing cells.

(Peters et al, 2015)

Study Design

Methodolgy: This is a Phase 1 multicenter, dose finding, open-label, single agent study of RX-3117 administered orally to subjects with advanced or metastatic solid tumors. One subject will be treated per dose group until the appearance of a related grade 2 or greater adverse event, afterwhich 3 subjects will be treated using the modified Fibonacci schedule.

Treatment: Subjects will be treated administered 3 times a week for 3 weeks followed by 1 week off in each 28 day cycle.

*

Sample Size

Approximately 30 subjects depending on the number of dose levels needed to identify the maximum tolerated dose (MTD)

Study Objectives

Primary objectives:
- Evaluate safety and tolerability of escalating doses of RX-3117 in subjects
- Determine the maximum tolerated dose of RX-3117

Secondary objective:
- Determine the safety and tolerability of multiple doses of RX-3117
- Determine the PK profile of RX-3117
- Evaluate the antitumor activity of RX-3117
- Evaluate the potential QT interval prolongation due to RX-3117

Exploratory objective:
- Investigate potential predictive and pharmacodynamic blood and tumor biomarkers

Study Endpoints

- **Primary endpoint:** Incidences of dose limiting toxicities (DLTs) in cycle 1
- **Secondary endpoint:** PK parameters and changes in tumor size
- **Exploratory endpoint:** Biomarker concentrations in blood

Key Inclusion Criteria

- Males or females ≥ 18 years old
- Histological or cytological evidence of confirmed solid tumor malignancy
- Able to discontinue all anticancer therapies 2 weeks prior to study start
- Measurable or evaluable disease using Response Evaluation Criteria in Solid Tumors
- Able to swallow capsules
- Life expectancy of at least 3 months
- ECOG performance status of 0, 1 or 2

Key Exclusion Criteria

- Primary brain tumors or clinical evidence of active brain metastasis
- Systemic corticosteroid use within 7 days before planned start of study therapy
- Active infection requiring parenteral or oral antibiotics within 2 weeks before planned start of study therapy
- Uncontrolled diabetes as assessed by the investigator
- Prior or current history of hepatitis B, hepatitis C or human immunodeficiency virus
- History of bone marrow of solid organ transplantation
- History of congestive heart failure, arrhythmias, acute coronary syndrome or torsades de pointes
- Any other medical, psychiatric, or social condition, which in the opinion of the investigator, would preclude participation in the study, pose an undue medical hazard, interfere with the conduct of the study, or interfere with interpretation of the study results
- Known hypersensitivity to gemcitabine, azacytidine or cytosine arabinoside
- Pregnant, planning a pregnancy or breast feeding during the study
- Concurrent participation in another therapeutic clinical trial

Reference

- Peters, G et al. AACR; 2015; Abstract nr 2622

Investigator Disclosures

- Dr. Drew Rasco – nothing to disclose
- Dr. Amita Patnaik – nothing to disclose
- Dr. Anthony Tolcher – nothing to disclose
- Dr. Christine Peterson – Rexahn Pharmaceuticals
- Dr. Ely Benaim – Rexahn Pharmaceuticals

For further information about RX-3117 and Rexahn Pharmaceuticals, Inc., please contact Dr. Christine Peterson: petersonc@rexahn.com, (240) 268-5300 x 320