A Phase 1 Study of RX-5902 (Supinoxin) an Oral Agent Targeting Phosphorylated p68 to Treat Subjects with Advanced Solid Tumors.

S. Gail Eckhardt[1], W. Larry Gluck[2], Martin Gutierrez[3], Christine Peterson[4] and Ely Benaim[4],
[1]University of Colorado, Anschutz Cancer Center, Aurora, CO, [2]Translational Oncology Research, Greenville, SC,
[3]John Theurer Cancer Center, Hackensack University Medical Center, Hackensack, NJ [4]Rexahn Pharmaceuticals, Inc., Rockville, MD

Abstract TPS#2608

Background: RX-5902 is a novel compound that targets phosphorylated p68 RNA helicase (also known as DDX5), a member of the DEAD box family of RNA helicases. Phosphorylated p68 may play a vital role in cell proliferation and tumor/cancer progression. As a single agent, RX-5902 inhibits tumor growth and enhances survival in a variety of in vivo animal xenograft tumor models (e.g., renal, ovarian, pancreatic, melanoma).

Methods: This Phase 1, open-label, multicenter study evaluates the efficacy and safety of RX-5902 in subjects with solid tumors. RX-5902 is administered orally once weekly for 3 weeks with 1 week of rest in each 4 week cycle. Dose escalation starts with an accelerated design treating 1 subject per dose followed by a standard 3 + 3 design using a modified Fibonacci sequence after the occurrence of a single Grade 2 or greater adverse event that is considered at least possibly related to RX-5902. The primary endpoint is the overall safety profile characterized by the type, frequency, severity, timing of onset, duration and relationship to study therapy of any adverse events, or abnormalities of laboratory tests or electrocardiograms as well as the description of any dose limiting toxicities that occur during Cycle 1, serious adverse events, or adverse events leading to discontinuation of study treatment. Secondary endpoints include pharmacokinetic parameters and Indices of anti-tumor activity. Exploratory endpoints are biochemical levels of drug targets in blood and tumor samples. Eligible subjects must have confirmed histologic or cytologic evidence of metastatic or locally advanced solid neoplasm that has failed to respond to standard therapy, progressed despite standard therapy or for which standard therapy does not exist. There is no limit on the number of prior treatment regimens.

NCT020030928

Proposed Mechanism of Action

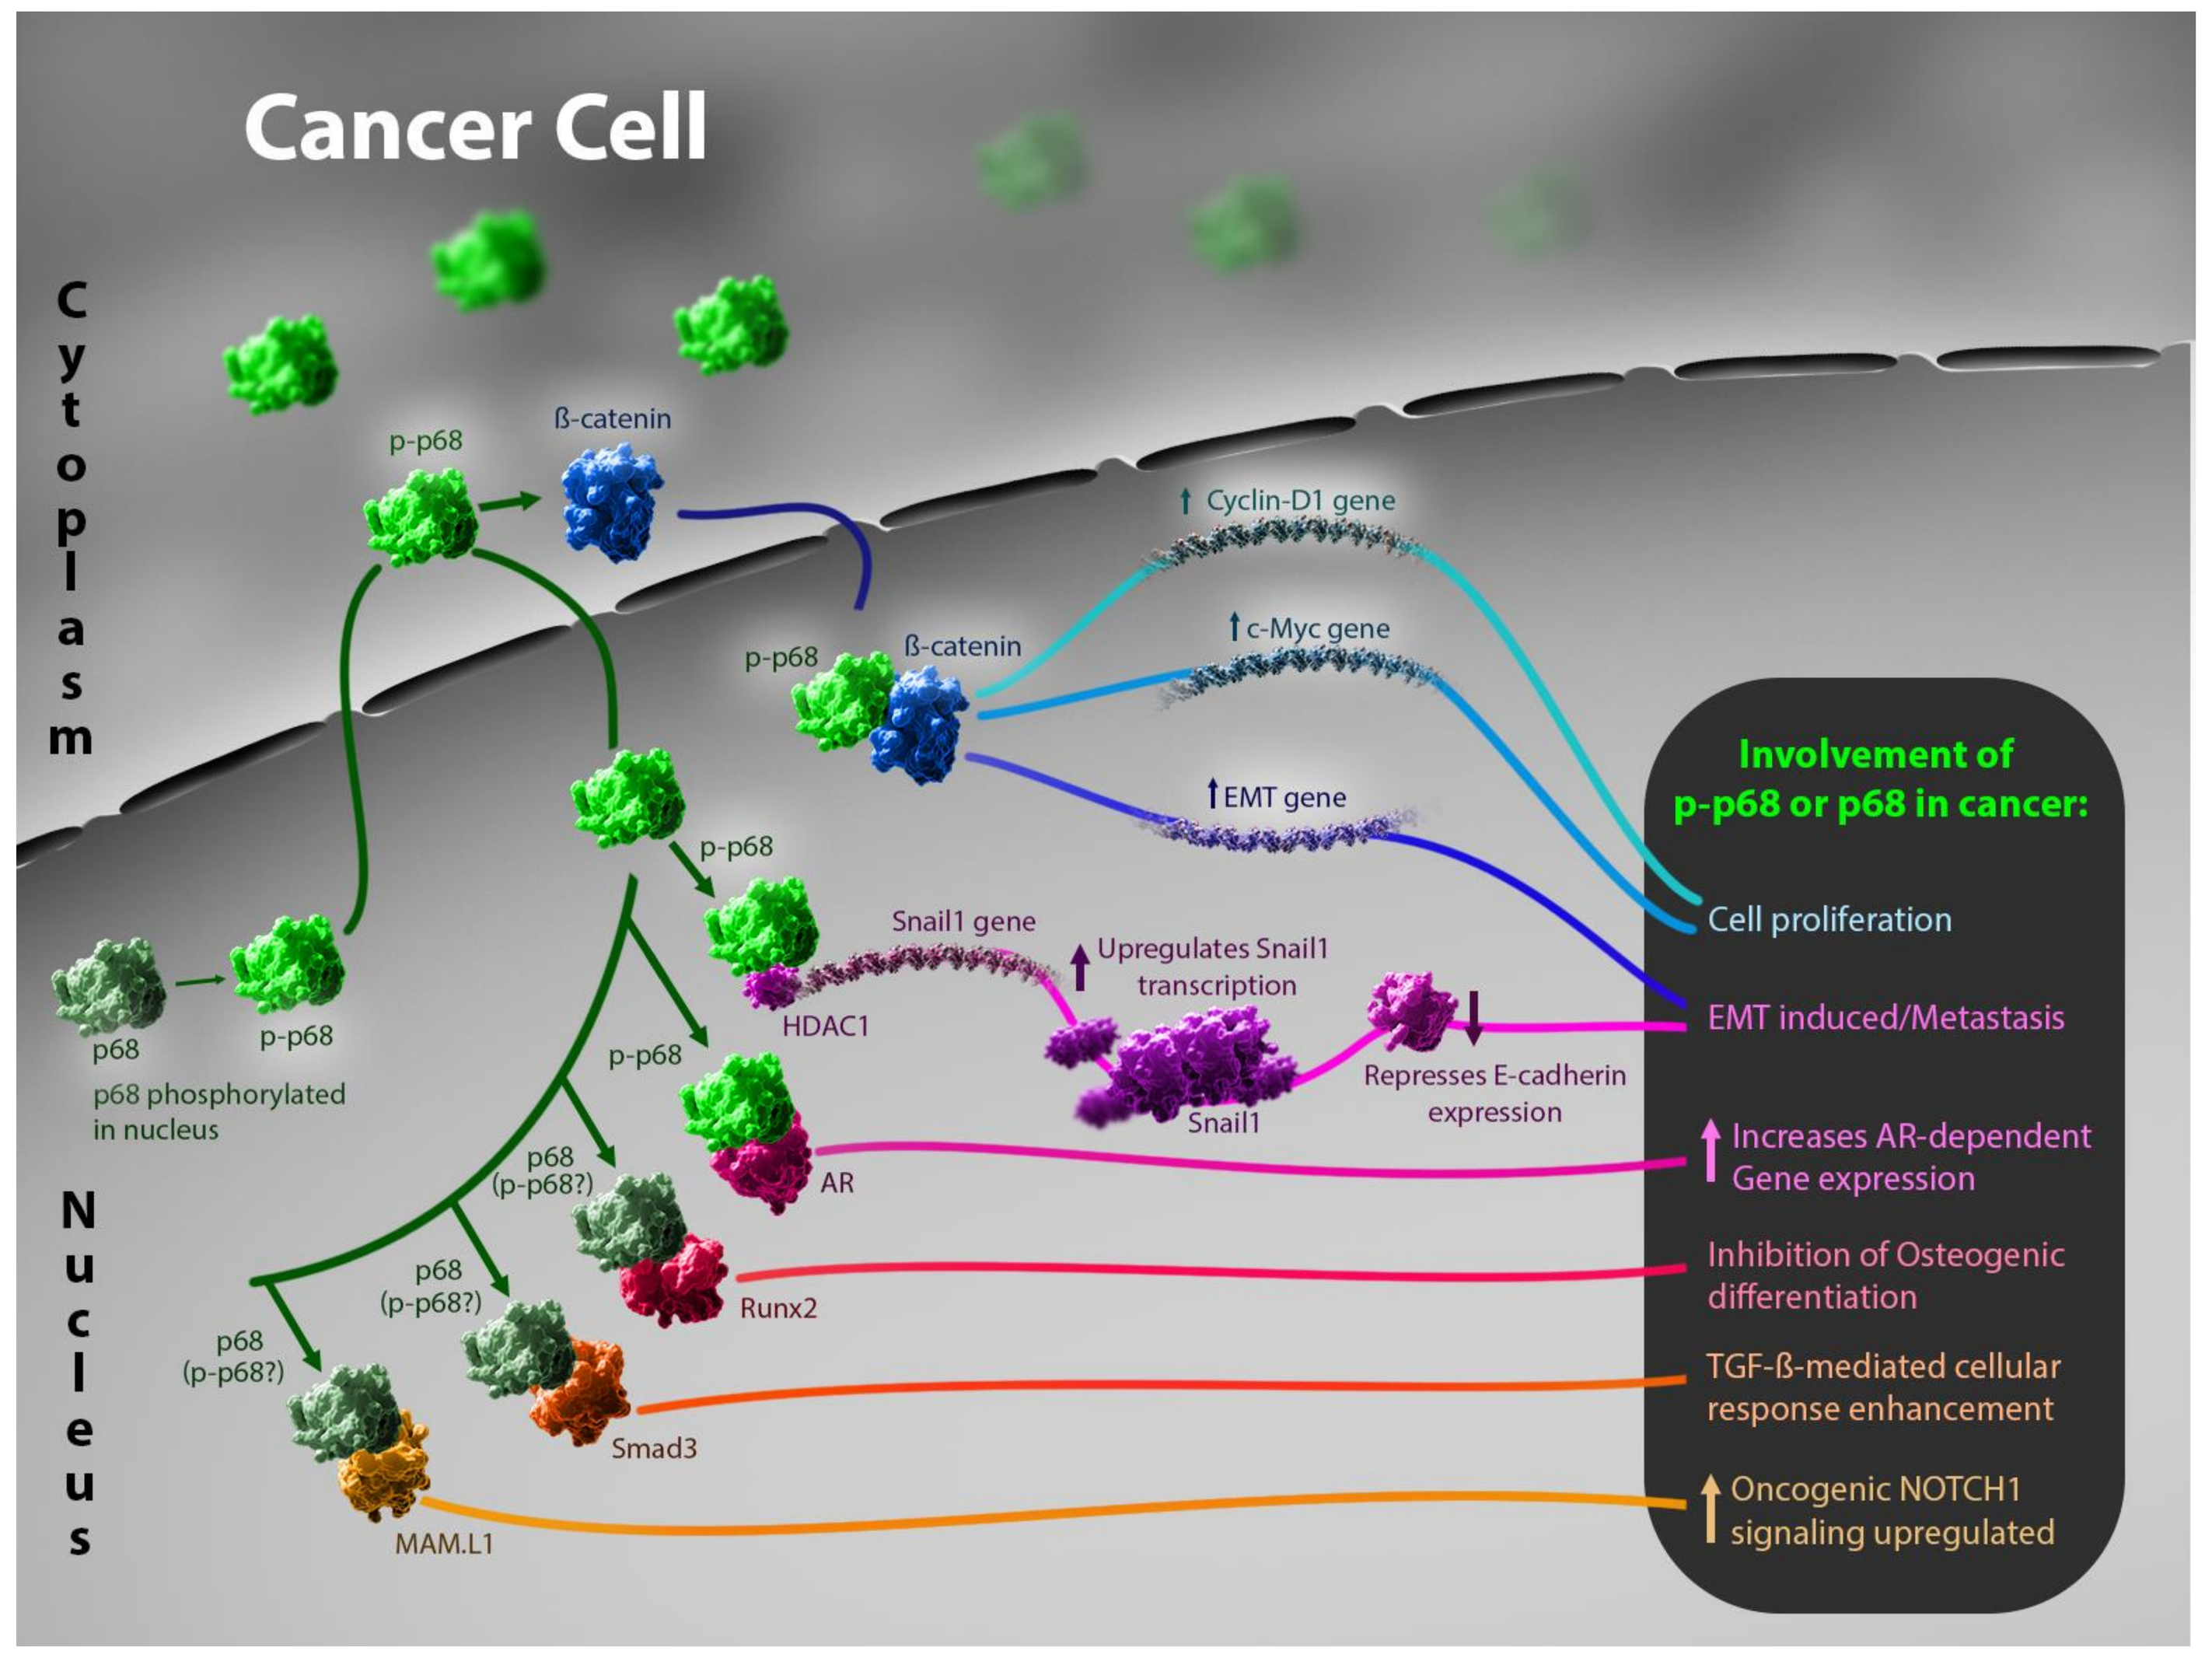
Cancer Cell
Cytoplasm
Nucleus
p-p68
ß-catenin
p-p68
ß-catenin
Cyclin-D1 gene
c-Myc gene
EMT gene
p-p68
Snail1 gene
Upregulates Snail1 transcription
HDAC1
Snail1
Represses E-cadherin expression
p68
p-p68
p68 phosphorylated in nucleus
p-p68
AR
p68 (p-p68?)
Runx2
p68 (p-p68?)
Smad3
p68 (p-p68?)
MAM.L1
Involvement of p-p68 or p68 in cancer:
Cell proliferation
EMT induced/Metastasis
Increases AR-dependent Gene expression
Inhibition of Osteogenic differentiation
TGF-ß-mediated cellular response enhancement
Oncogenic NOTCH1 signaling upregulated

Pancreatic Cancer Model of Anti-tumor Activity

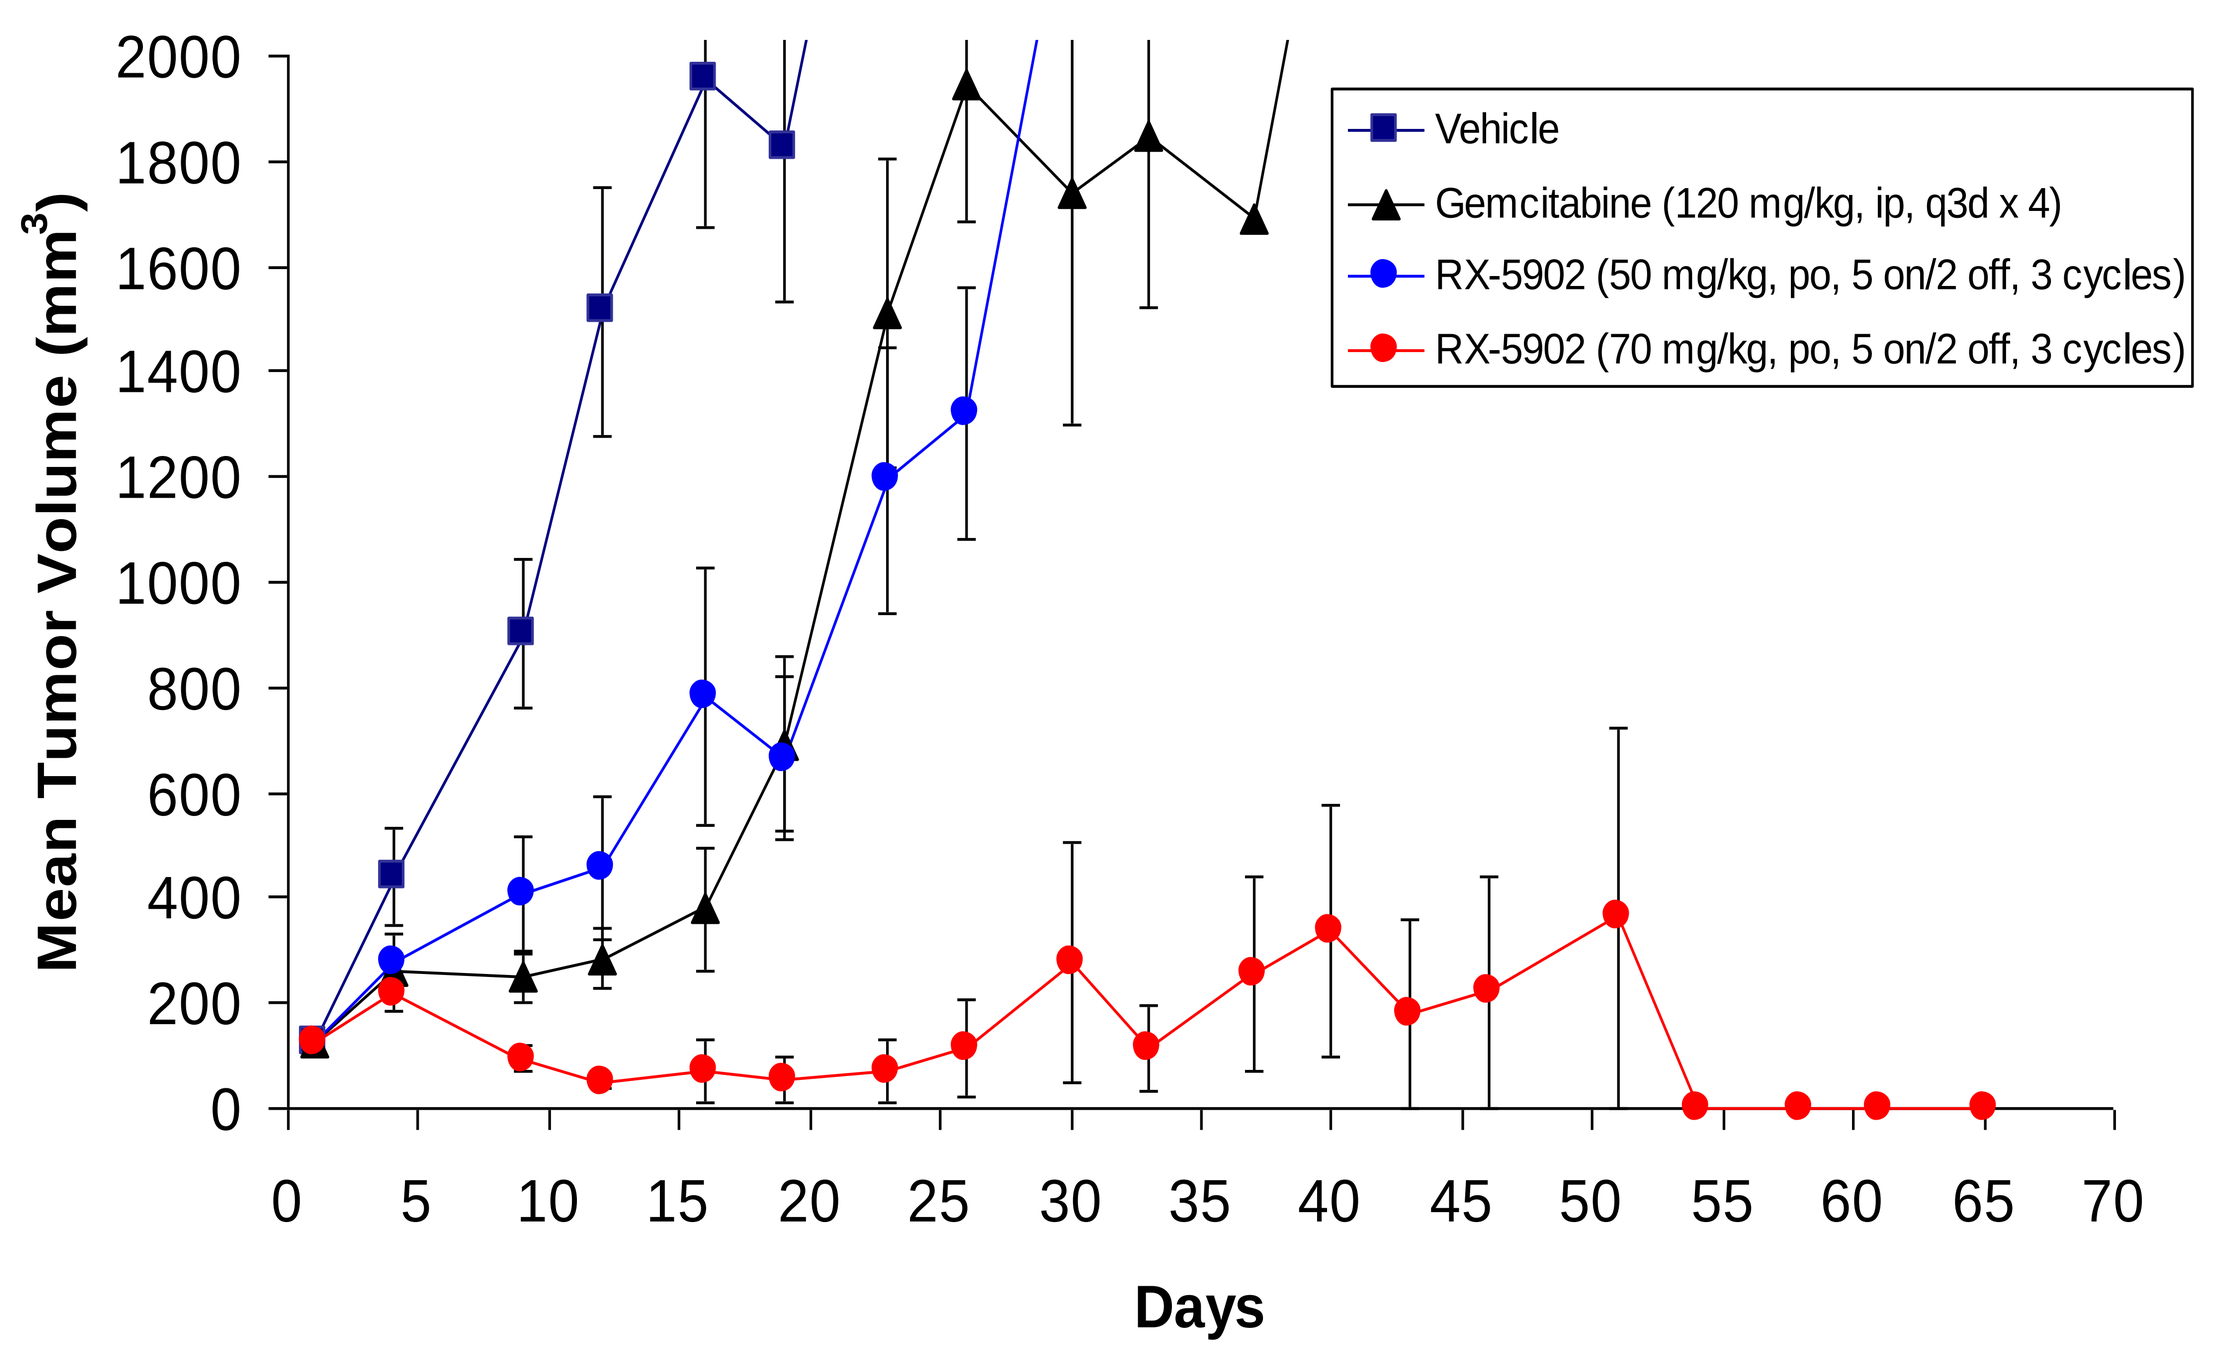
Mean Tumor Volume (mm^3)
Days
0
200
400
600
800
1000
1200
1400
1600
1800
2000
0 5 10 15 20 25 30 35 40 45 50 55 60 65 70
Vehicle
Gemcitabine (120 mg/kg, ip, q3d x 4)
RX-5902 (50 mg/kg, po, 5 on/2 off, 3 cycles)
RX-5902 (70 mg/kg, po, 5 on/2 off, 3 cycles)

Melanoma Model of Anti-tumor Activity

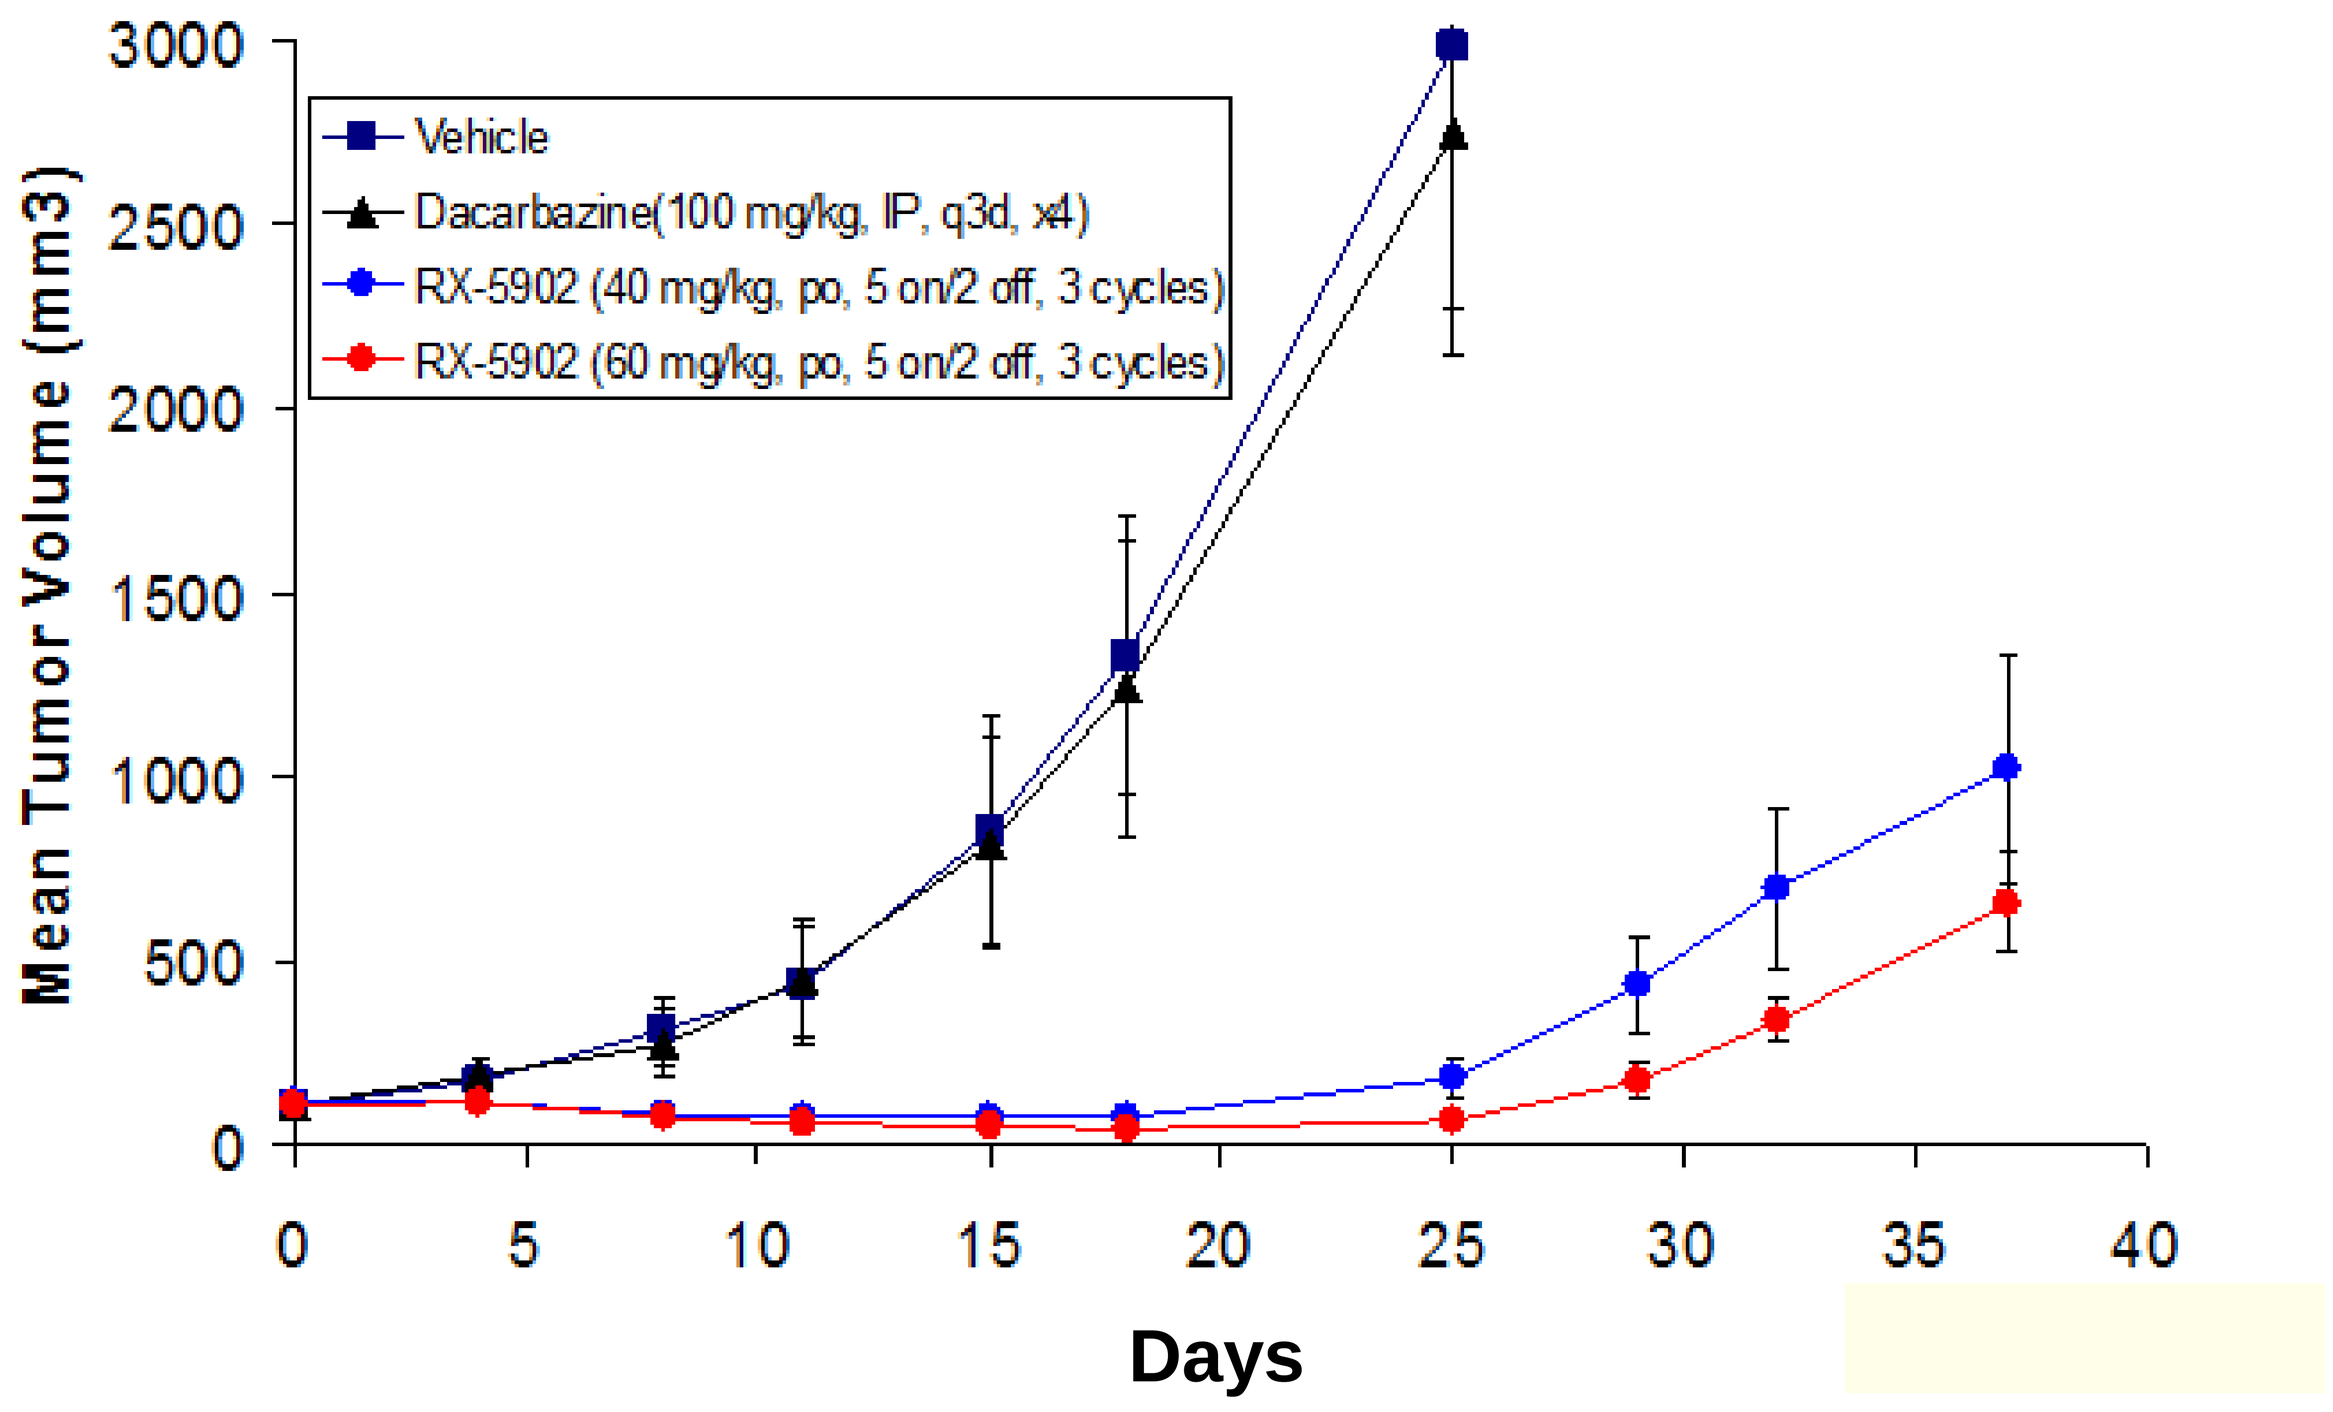
Mean Tumor Volume (mm3)
Days
0
500
1000
1500
2000
2500
3000
0 5 10 15 20 25 30 35 40
Vehicle
Dacarbazine(100 mg/kg, IP, q3d, x4)
RX-5902 (40 mg/kg, po, 5 on/2 off, 3 cycles)
RX-5902 (60 mg/kg, po, 5 on/2 off, 3 cycles)

Study Design

Methodology: This is a Phase 1 multicenter, dose finding, open-label, single agent study of RX-5902 administered orally to subjects with advanced or metastatic solid tumors. One subject will be treated per dose group until the appearance of a related grade 2 or greater adverse event, after which 3 subjects will be treated using the modified Fibonacci schedule.

Treatment: Subjects will be treated for up to 6 cycles of therapy. RX-5902 will be taken on Days 1, 8, and 15 followed by a week of rest on Day 22 of a 28-day cycle. All subjects will be followed for at least 30 days after the last dose of RX-5902. Additional cycles allowed for subjects receiving benefit.

Study Objectives

Primary objectives:
- Evaluate safety and tolerability of escalating doses of RX-5902 in subjects

Secondary objectives:
- Characterize the safety and tolerability of multiple doses of RX-5902
- Characterize the PK profile of RX-5902
- Characterize the antitumor activity of RX-5902

Exploratory objective:
- Investigate the effect of RX-5902 on potential biomarkers (e.g., P-p68, c-myc, c-jun) in blood or tumor samples

Key Inclusion Criteria

- Male and female patients who are 18 yrs or older
- Histologically confirmed solid tumor malignancy that is refractory, intolerant, or ineligible to receive approved standard therapy
- Measurable or evaluable disease per Response Evaluation Criteria in Solid Tumors (RECIST)
- Able to swallow capsules

Key Exclusion Criteria

- Primary brain tumor or active brain metastases
- Persistent toxicities >Grade 1 (except Grade 2 alopecia or neuropathy) associated with previous cancer therapies
- Any other cancer treatment within 2 weeks of planned study treatment
- History of clinically significant GI bleed, intestinal obstruction, or GI perforation within 6 months of study dosing
- History of long QT syndrome or clinically significant cardiac arrhythmias (except stable atrial fibrillation)
- Myocardial infarction within 6 months of study dosing
- Active infection requiring IV antibiotics within 2 weeks of dosing
- History of Hepatitis B, C, or HIV
- Use of potent inhibitor or inducer of CYP3A4/3A5 within 14 days of planned study treatment or expected requirement for use of such a drug during study
- Use of a potent inhibitor or inducer of drug transporters or conjugating enzymes within 14 days prior to planned study treatment or expected requirement for use of such a drug during study

References

- Yang et al. (2006) Cell, 127: 139-155.
- Kost et al. (2015) J. Cell. Biochem. 10.1002.
- Lee et al, AACR; Cancer Res 2011;71(8 Suppl):Abstract nr 1371

Investigator Disclosures

- Dr. S. Gail Eckhardt– nothing to disclose
- Dr. W. Larry Gluck – nothing to disclose
- Dr. Martin Gutierrez– nothing to disclose
- Dr. Christine Peterson – Rexahn Pharmaceuticals
- Dr. Ely Benaim – Rexahn Pharmaceuticals

For further information about RX-5902 and Rexahn Pharmaceuticals, Inc., please contact Dr. Christine Peterson: petersonc@rexahn.com, (240) 268-5300 x 320